EULAR14-SCIE-5515

Creating New Rheumatologists: The Canadian Experience

A. Cividino[1], S. Barr[2], V. Bakowsky[3], L. Bessette[4], J. Homik[5], N. Khalidi[1], C. Pineau[6], J. Pope[7], D. Robinson[8], K. Shojania[9], E. Yacyshyn[5]

[1]McMaster University, Hamilton; [2]University of Calgary, Calgary; [3]Dahousie University, Halifax; [4]Laval Université, Québec; [5]University of Alberta, Edmonton; [6]McGill University, Montréal; [7]Western University, London; [8]University of Manitoba, Winnipeg; [9]University of British Columbia, Vancouver

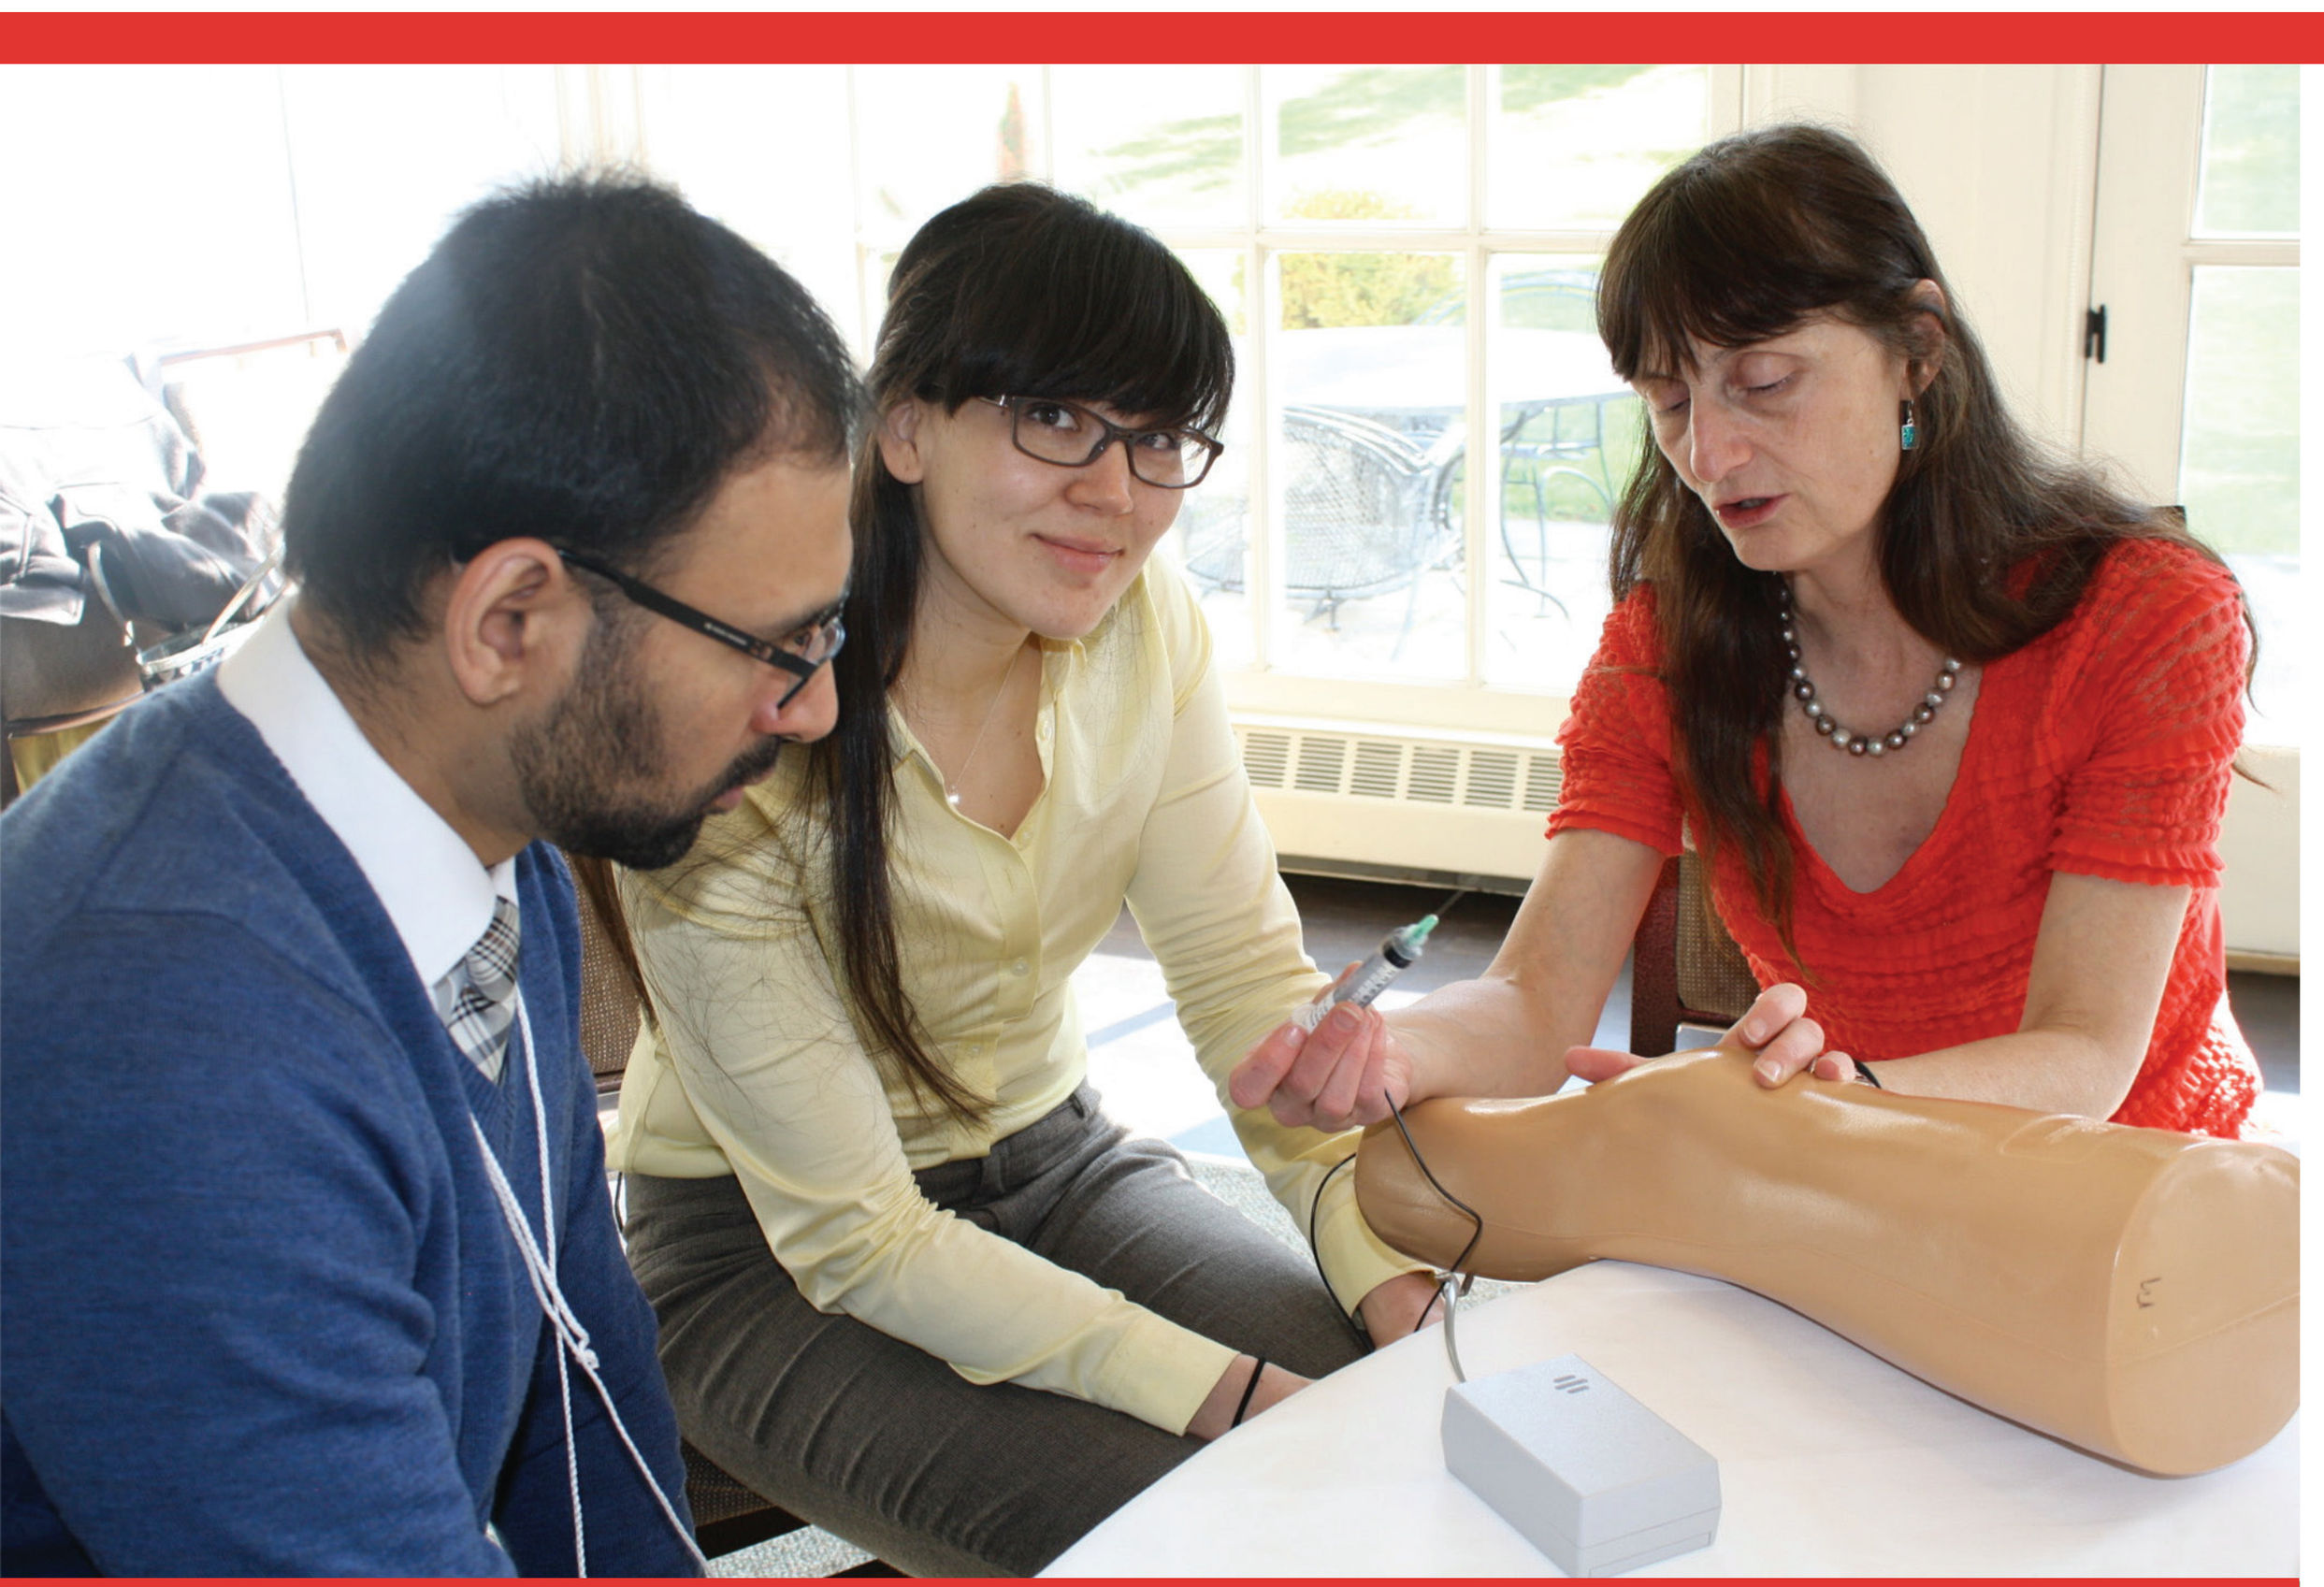

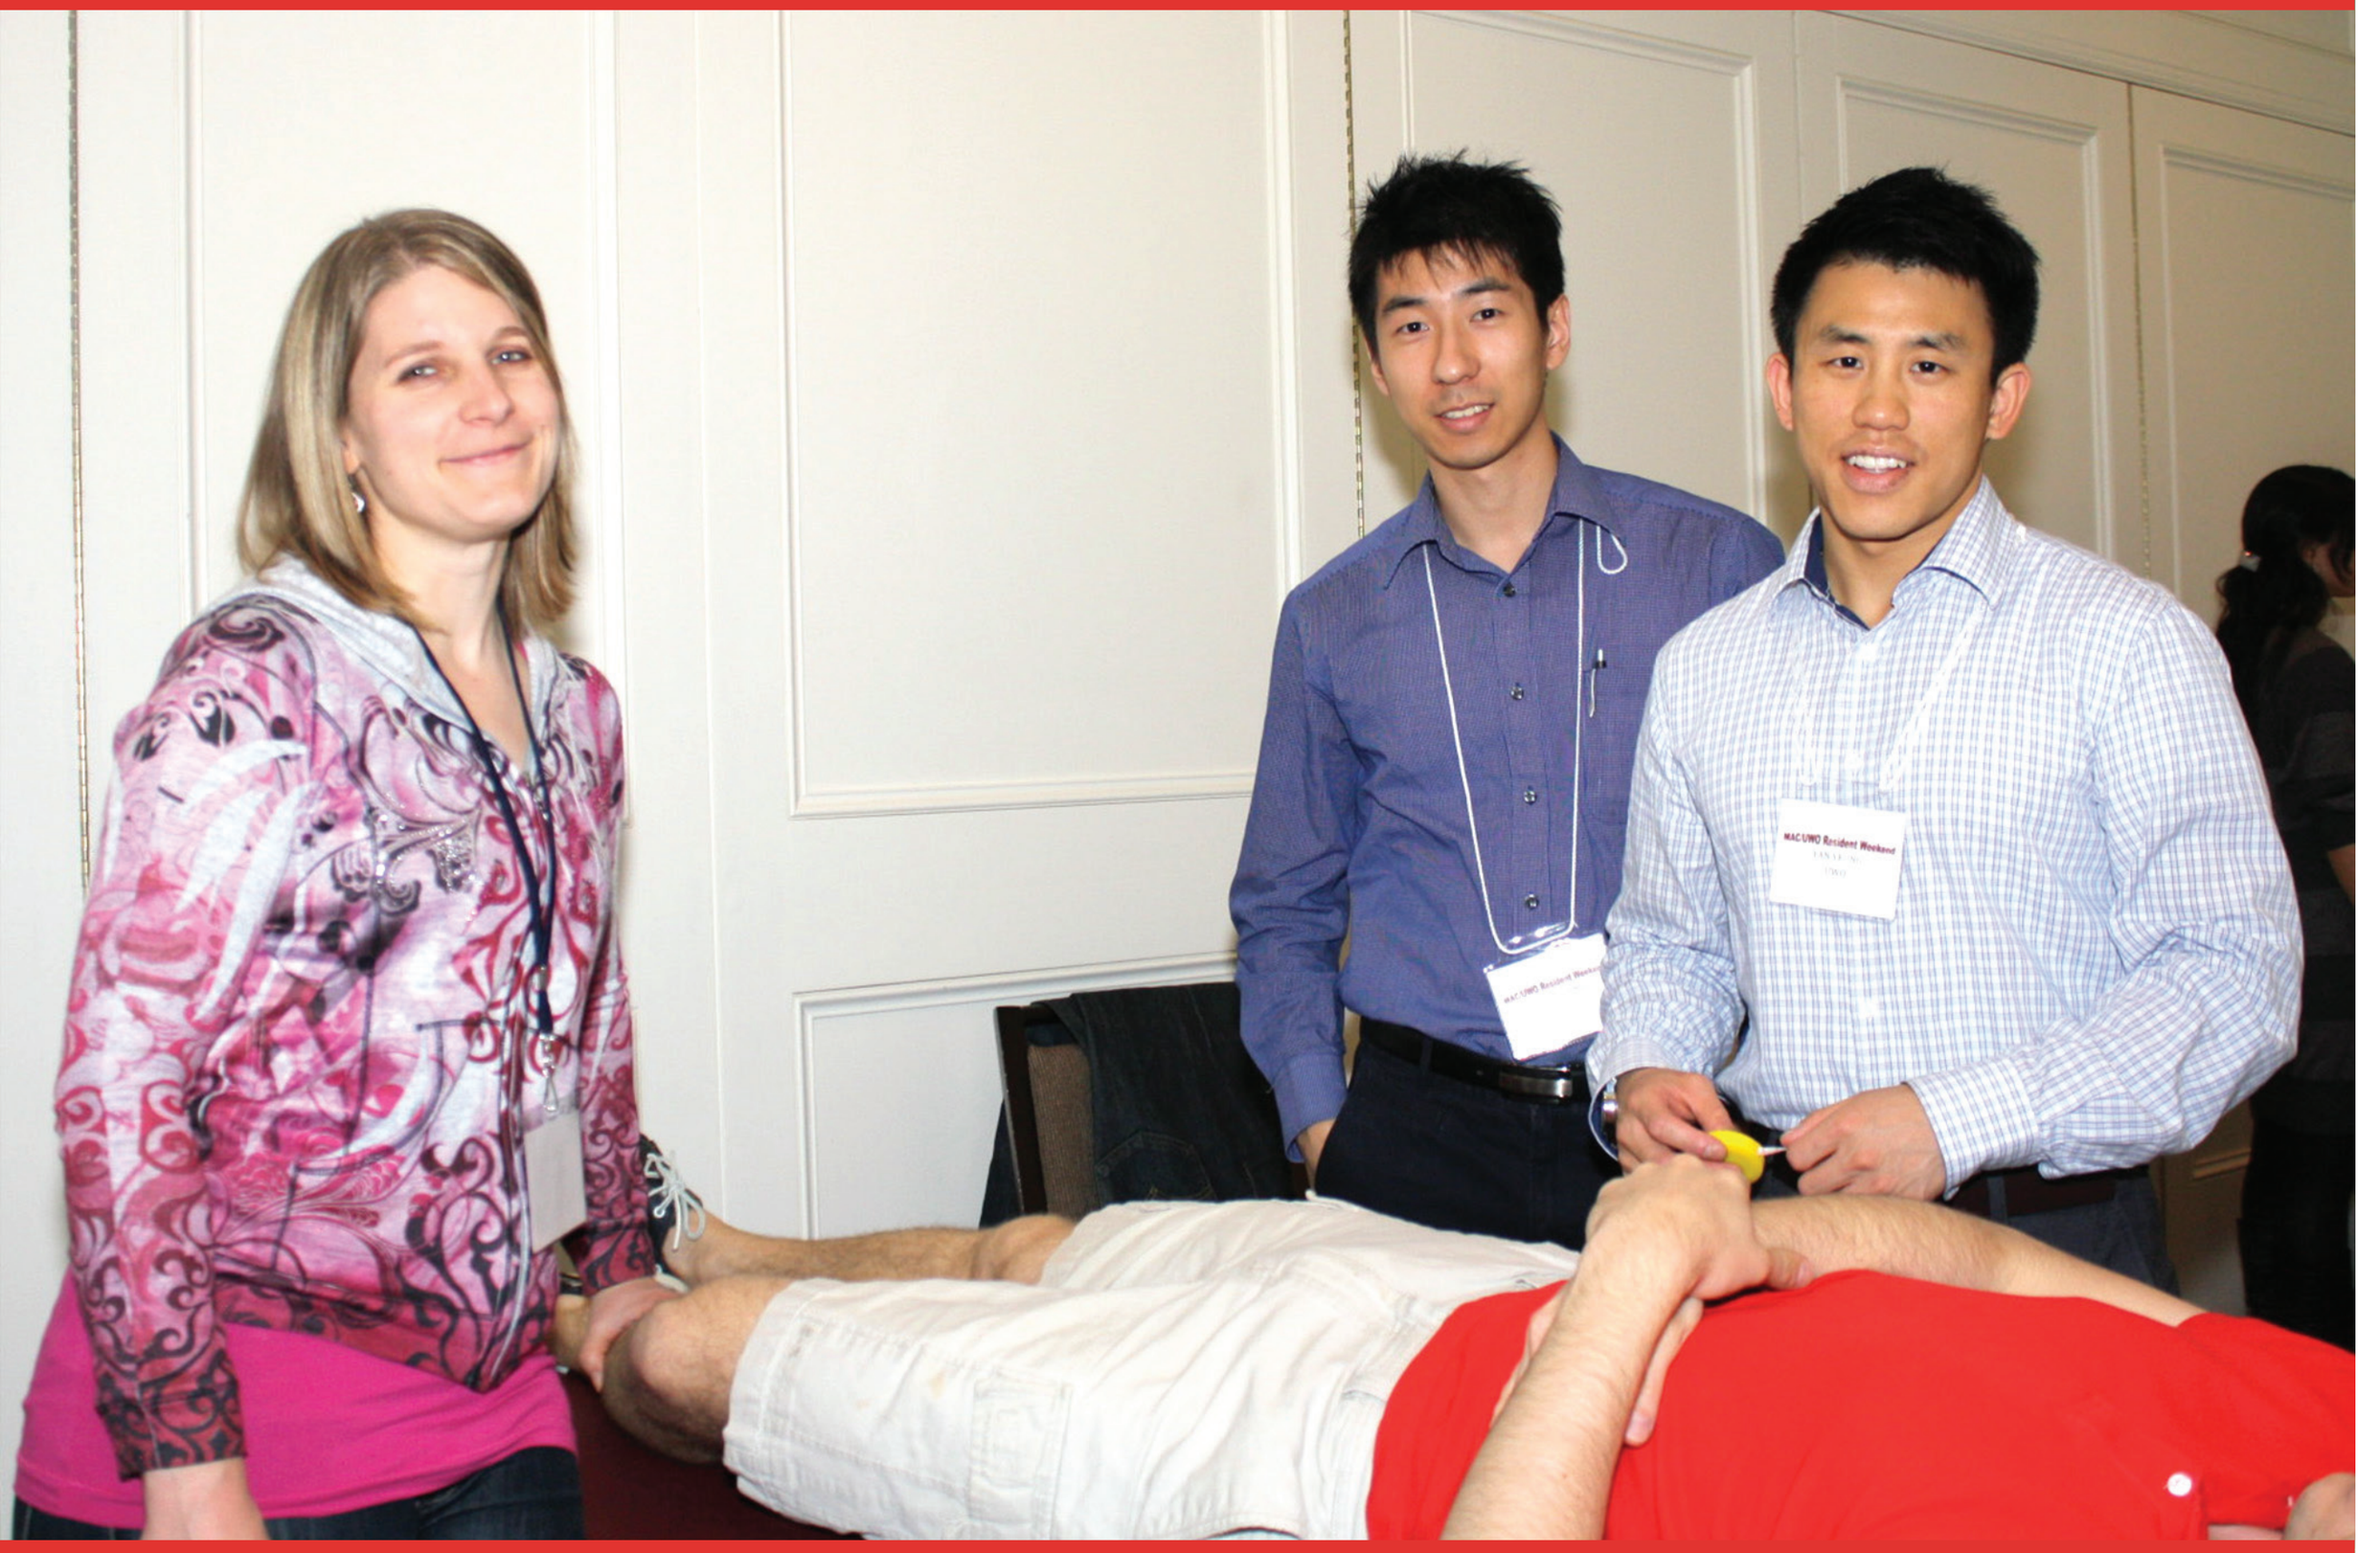

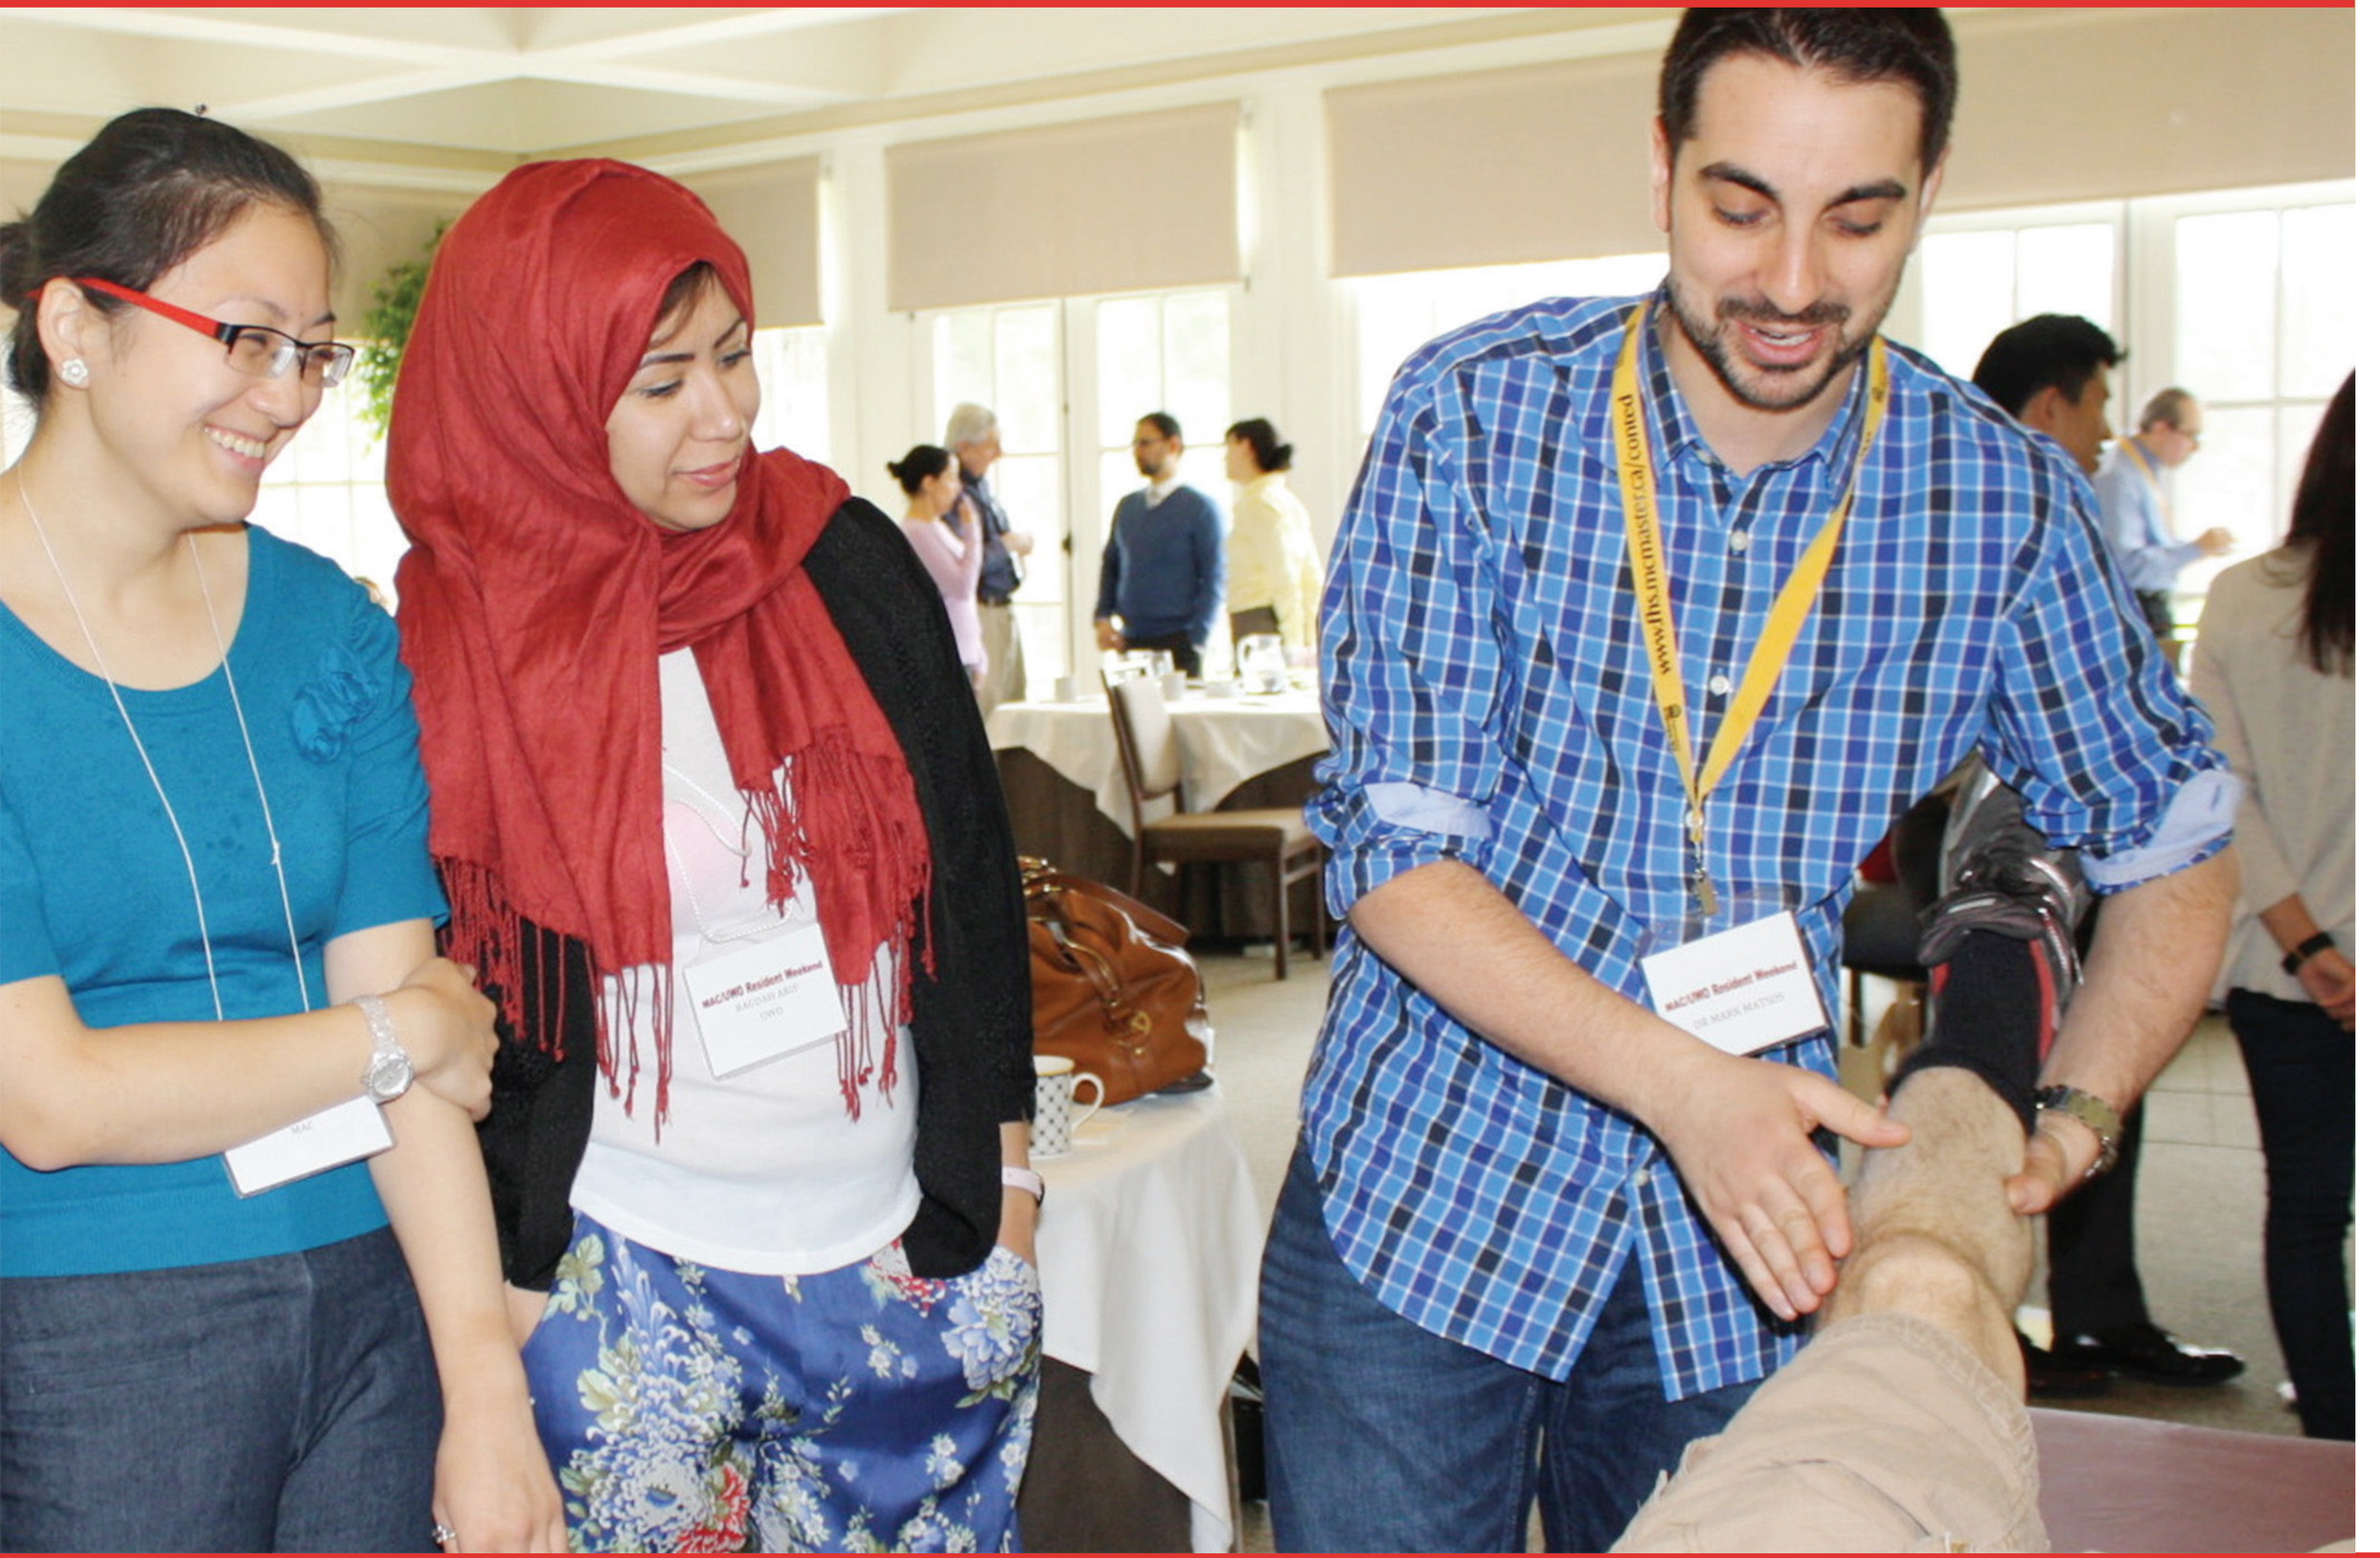

BACKGROUND

Many countries face a critical shortage of rheumatologists. Based on an accepted benchmark of 1 specialist per 50,000 people as the number needed for effective patient care,[1] recent figures show shortages in some EU Member states,[2] parts of the US[3] and in Canada.[4] This qualitative environmental scan was designed to identify how Canadian rheumatologists are trained, perceived positive aspects of a career in this subspecialty, how programs inform/attract learners and what can be done collectively to attract more trainees.

METHODS

Data Collection:

Rheumatology programs across Canada were asked to send invitation emails to their faculty and trainees. Interested parties could complete a self-administered online survey in English or French, or take part in an individual telephonic interview in English.

Data Analysis:

Individual-level data from program faculty, administrators and learners across Canada (n = 103, 78 surveys, 25 interviews). We found no major differences between research sites and so combined the data during analysis. Survey data were subjected to Thematic Framework Analysis to identify key concepts and issues from all sites. This was followed by a careful review of the transcripts to identify further examples or new concepts. Information on what attracted respondents to rheumatology and appropriate messaging differed by respondent group, learners (undergraduate medical students, junior residents [PGY1-3], senior residents [PGY4-6]) and faculty/administrators (division and program directors, faculty and laypersons involved with a rheumatology program), and so were analyzed separately.

RESULTS

Participants:

Division and Program Directors of nine post-graduate rheumatology programs across Canada collaborated on this project. There were 103 respondents, 52 learners and 51 faculty/ administrators. Most respondents were female (67% of survey responders and 55% of interviewees).

Ways to Increase Interest in Rheumatology:

The need to train more rheumatologists is widely recognized. Respondents expressed an overall enthusiasm about the profession (*"La rhumatologie est la spécialité d'avenir au Canada"* [L]). Rheumatologists on faculty noted the need to change the face of rheumatology (*"We need to get rid of the label 'arthritis centres'. Rheumatology is often thought of as dealing only with arthritis and diseases of the elderly. We need to* [instead] *promote rheumatology as a specialty with fascinating immunopathogenesis and cutting edge research at the frontier of discovery."* [F]). Respondents advocate targeting both undergraduates (*"People who influenced me were* [role models] *I had as a medical student"* [F]) and junior Internal Medicine residents. Most recommendations focused on ways to increasing exposure to the field through face-to-face interactions with rheumatologists, courses in medical school curricula, and specific messages to enhance knowledge of and interest in the field of rheumatology (*"Tell them it exists; I didn't know about rheumatology until the start of Internal Medicine"*[L]); *"Emphasize happy doctors"*, [L]).

Junior learners suggested personal contact with practitioners through career talks, observerships, shadowing and mentoring opportunities for undergraduates, as well as information sessions in small groups such as during lunch or dinner meetings where rheumatologists describe their careers. They also thought that formal courses in medical school such as in an MSK block or clinical skills sessions were important (*"More teaching by rheumatologists in the medical curriculum so students are aware of what the specialty is"* [L]).

Senior learners also noted the importance of early exposure to rheumatology for undergraduates rather than focusing efforts on reaching Internal Medicine residents. The need for adequate rotation placements was also highlighted (*"Make sure that we're not turning away interested Internal Medicine residents because we've taken on too many other kinds of residents for electives."* [F])

Messages to brand rheumatology as an attractive specialty included the intellectual challenge (*"This field fascinates me"* [L]; *"novel immunotherapies make it very exciting"* [F]), the stimulating workday (*"nice mix of procedural and cerebral work"* [L]), positive relationships with colleagues (*"According to a recent survey we are the happiest specialists"* [F]), alleviating suffering (*"We know how to treat arthritis now"* [L]), good quality of life (*"excellent work-life balance"* [F]) and excellent job prospects (*"the health care system needs you"* [F]). They also suggested telling students about widespread availability of jobs in the field.

CONCLUSIONS

This is the first cross-Canada qualitative study providing the insiders' view on what is needed to attract more trainees to the field of rheumatology. We found consensus on the need to inform potential trainees about rheumatology early in their education through a variety of messages and methods. Because of the shortage of rheumatologists it is important to increase awareness and information about the field by selectively using limited resources. This points to a need to collaboratively develop tools and methods to increase interest in rheumatology for use in multiple contexts.

Key to Quotes: [L] = learner; [F] = faculty.

References: [1]Karolinska Institutet/i3 Innovus. (2009) (http://www.comparator- reports.se/RA%20 Barrier%20Report_FINAL_050110.pdf); [2]Ibid.; [3]Deal et al. (2007). *Arthritis & Rheumatism* 56(3): 722-729; [4]2012 CMA Masterfile (www.cma.ca /multimedia/CMA/Content_Images/Inside_cma/Statistics/ 02SpecAge.pdf)

Photos taken by Rennée Tremblay, coordinator for the jointly hosted 10th annual McMaster/UWO PGY 1 & 2 Internal Medicine Residents Weekend in Rheumatology. The weekend gives internal medicine trainees with an interest in rheumatology the opportunity to spend time with faculty as well as practice examination stations, Royal College exam scenarios and OSCE exams.